A Dose-Finding and Phase II Study of selumetinib (Hyd-Sulfate), AZD6244, in Combination with Irinotecan as 2nd Line Therapy in Patients with KRAS or BRAF Mutation Positive Advanced Colorectal Cancer

H. S. Hochster[1], S. G. Groshen[2], W. A. Messersmith[3], S. G. Eckhardt[3], P. D. Smith[4], G. Y. Locker[5], G. Orphanides[4], L. P. Leichman[6]

Aptium GI Cancer Consortium (AGICC);

Yale Cancer Center, New Haven, CT[1]; University of Southern California and Norris Comprehensive Cancer Center, Los Angeles, CA[2]; University of Colorado Cancer Center, Aurora, CO[3]; AstraZeneca, Cheshire, England[4]; AstraZeneca, Wilmington, DE[5]; Comprehensive Cancer Center at Desert Regional Medical Center, Palm Springs, CA[6]

Abstract

Background: The MEK inhibitor selumetinib (ARRY-142886) is a potent inhibitor of MEK (MAP kinase kinase) with IC50 of 10-14 nM. This agent shows preclinical activity in tumors with activated EGF pathway signaling, particularly in cells with BRAF or KRAS mutation. A randomized phase II study compared this agent alone to capecitabine in previously treated CRC patients; it showed similar activity to the chemotherapy. We postulated selumetinib was most likely to show activity in patients with activating mutations of KRAS (codons 12 &13), shown unresponsive to anti-EGFR antibodies, or BRAF (V600E). We chose to add selumetinib to irinotecan as 2nd line therapy, as no other options beyond single agent irinotecan (or FOLFIRI) are available.

Methods: In this 2nd line study patients with advanced CRC and documented mutation of KRAS or BRAF, previously treated with one prior oxaliplatin-fluoropyrimindine plus bevacizumab regimen are eligible. Measurable disease and normal organ function are required. Patients are treated with irinotecan (180 mg/m2 q2w) and escalating doses of selumetinib in the phase I portion of this study (50 to 75 mg bid) in a 3+3 design, followed by a phase II study at the optimal dose combination for a total of up to 57 patients. Patients will undergo limited PK sampling for selumetinib and metabolites and tissue collection of PEFFT for EGFR, pEGFR, ERK, pERK, and topo-1 expression. A gene array profile is being developed to investigate MEK inhibition. Tumor biopsies will be performed in a subset of patients at baseline & day 15 (±3). Primary endpoint is response rate (RR). Compared to a historical control of 4% in the EPIC trial, a response rate greater than 15% in this setting would be promising for further drug development. A Simon two-stage design will be used for early stopping (≤1/20 and ≤4/45 responses), with 90% power to correctly conclude RR ≥15% and 10% chance of falsely concluding a RR of ≤4% .

Results: The study began accruing patients in May 2010. Results of the initial phase I patients will be presented.

Aptium Oncology
GI Cancer Consortium

I. AZD6244 (ARRY-142886): a MEK inhibitor

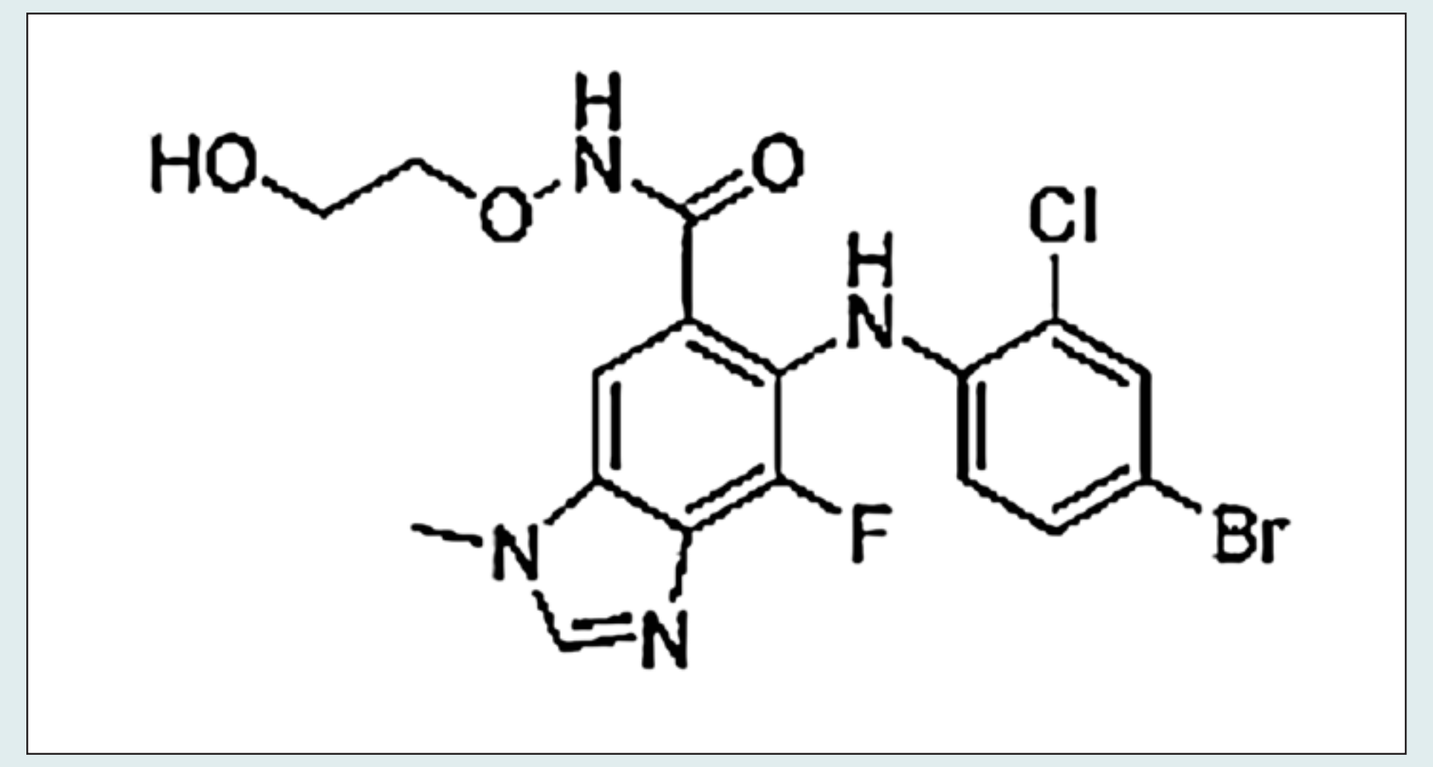

HO
O
N
H
O
H
N
Cl
F
Br
N
N

M.Wt: 457.68
Formula: C17H15BrClFN4O3
Solubility: 20 mmol/L in DMSO

*Data for "mixed drink" base compound shown, not Hyd-sulfate

II. Rationale

- Selumetinib small molecule uncompetititve inhibitor of MEK
- Potent preclinical activity in cell lines and xenografts with KRAS mutations
- Randomized single agent phase II studies
 - Selumetinib vs. cape in CRC (2 prior rx) – PFS equivalent;
 - **RR = 10 SD (v 1PR, 15 SD)**
 - Selumetinib vs. pemetrexed in NSCLC
- Combination studies
 - Phase II with docetaxel 75 mg/m2;
 - **MTD AZD 6244 = 75 po bid**
 - Phase II with DTIC in melanoma (selected for KRAS and BRAF mutation), BRAF mutated melanoma; KRAS mutated NSCLC

III. IN VIVO PRE-CLINICAL ACTIVITY

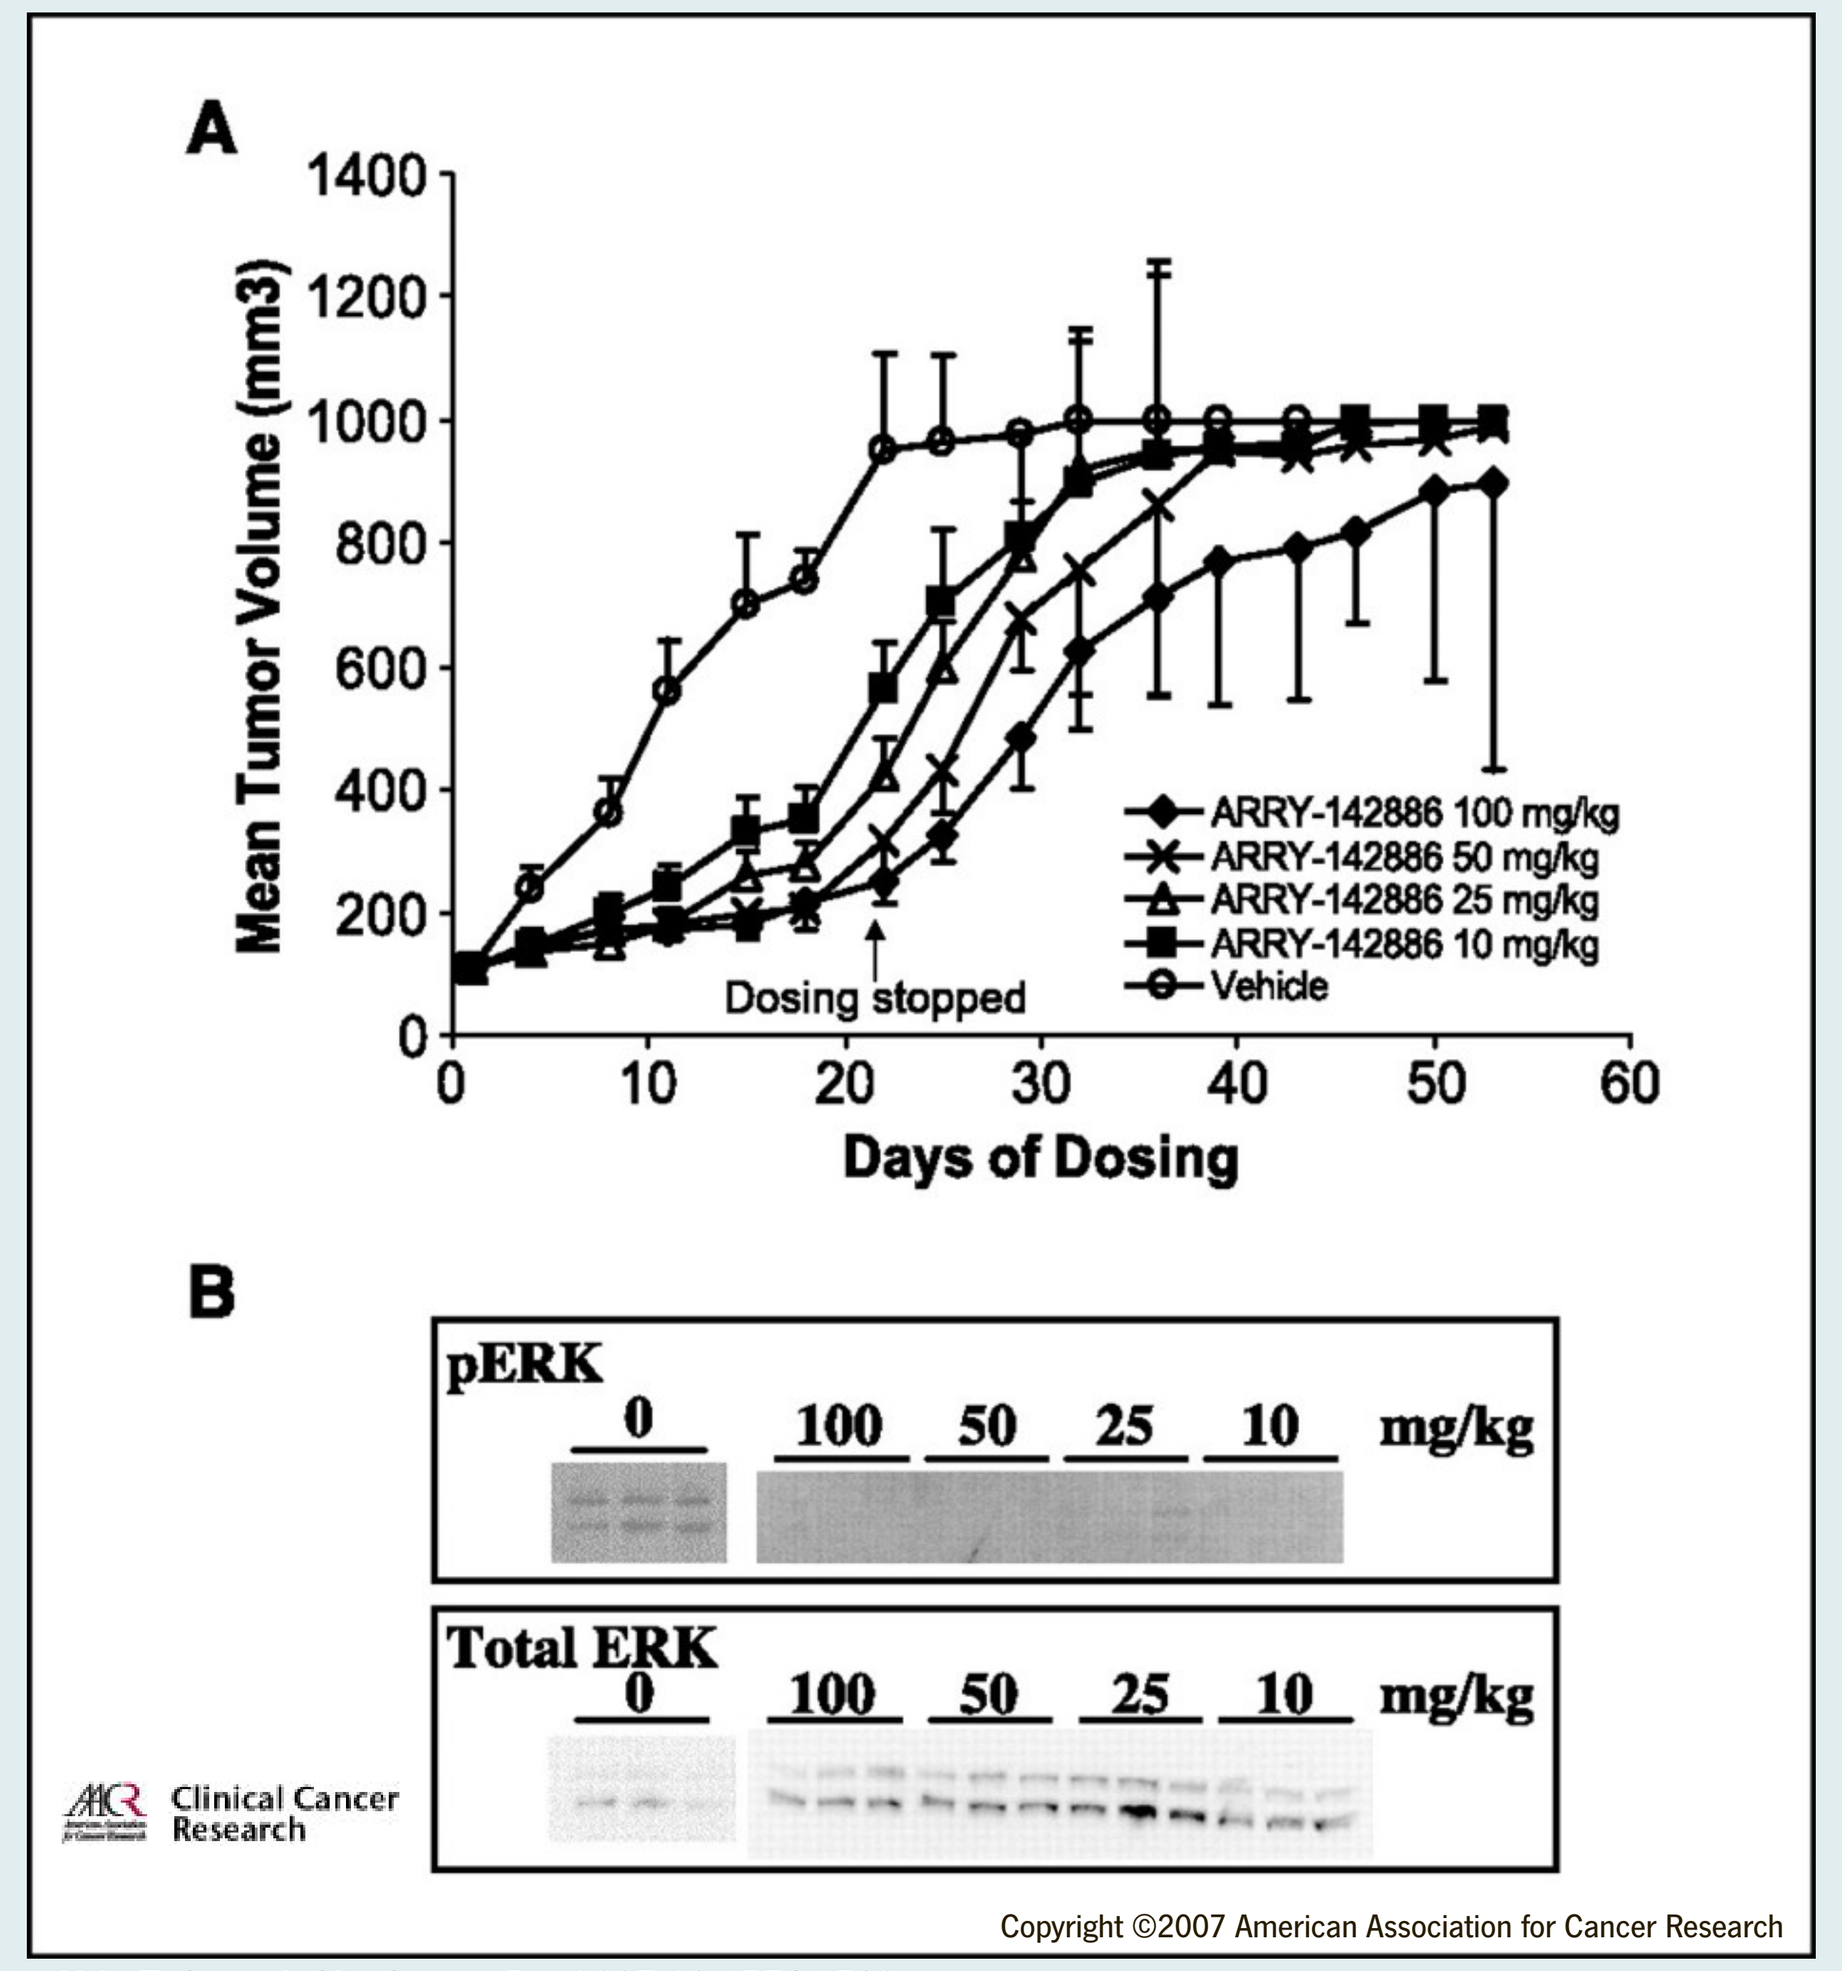

A
Mean Tumor Volume (mm3)
Days of Dosing
ARRY-142886 100 mg/kg
ARRY-142886 50 mg/kg
ARRY-142886 25 mg/kg
ARRY-142886 10 mg/kg
Vehicle
Dosing stopped
B
pERK
0 100 50 25 10 mg/kg
Total ERK
0 100 50 25 10 mg/kg
Clinical Cancer Research
Copyright ©2007 American Association for Cancer Research

Yeh, T. C. et al. Clin Cancer Res 2007;13:1576-1583

Inhibition of tumor growth and decreased tumor phospho-ERK1/2 levels in a mouse HT-29 xenograft model.

A, mice bearing HT-29 tumors were dosed with vehicle or selumetinib (10, 25, 50, or 100 mg/kg, oral, BID) for 21 d (n = 10 per group). Animals were sacrificed when tumors reached 1,000 mm3 in size. The larger error bars at end of study are due to the reduced number of animals left on study.

B, 4 h after the last dose on day 21, three mice per group were euthanized. Homogenates from excised tumors were analyzed for phospho-ERK1/2 and ERK1/2 expression by Western blotting.

IV. CRC: IN VIVO SENSITIVITY (data on File, AZ)

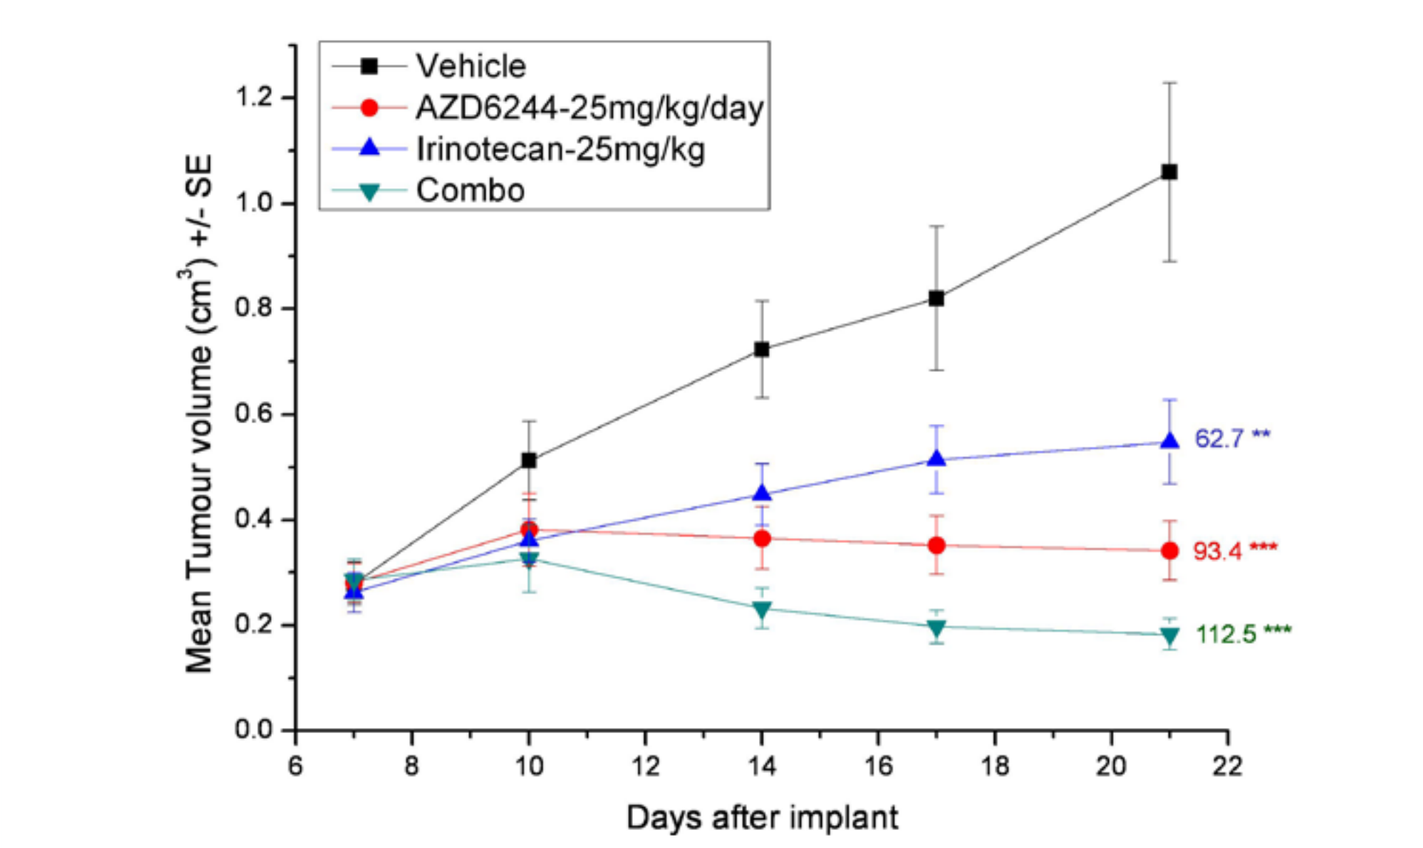

Vehicle
AZD6244-25mg/kg/day
Irinotecan-25mg/kg
Combo
Mean Tumour volume (cm3) +/- SE
Days after implant
62.7 **
93.4 ***
112.5 ***

HCT-116 colorectal tumor cells (1 x 107) were implanted into flank of female nude mice.

Tumors selected when they reached approximately 0.27 cm3 and randomised into treatment groups.

Selumetinib dosing commenced as indicated on the results graph on day 8 post-implantation and was continued p.o. twice daily until for 14 days; irinotecan was dosed i.v. once weekly on day 8 and day 15

V. CLINICAL Rationale

- Oxaliplatin-fluoropyrimidine with bevacizumab regimens are standard 1st line therapy for advanced CRC
- Irinotecan as second line therapy has consistently shown a PFS or 2.5 months and RR of 4%
 - (refs: Tournigand, et.al. JCO 22:229,2004 and Sobrero, et. al. [EPIC] JCO 26:2311,2008)
- Patients with KRAS or BRAF mutations do not benefit from anti EGFR antibodies
- Selumetinib, a MEK inhibitor, may particular be active in those patients with activating KRAS or BRAF mutations
- A small randomized phase II study showed single agent selumetinib equally active to capecitabine in previously treated CRC

VI. Eligibility: Inclusion

- Age 18 years or older
- Histological or cytological confirmation of CRC with available tissue, currently stage IV
- Failure of 1st line anti-cancer therapy with an oxaliplatin and bevacizumab based regimen (either radiological documentation of disease progression or due to toxicity) or subsequent relapse of disease following 1st line therapy; recurrence within 12 months of adjuvant FOLFOX acceptable
- ECOG Performance Status 0 – 1
- RECIST measurable: At least one lesion, not previously irradiated, that can be accurately measured as ≥10 mm by spiral computed tomography (CT) scan or as ≥20 mm with conventional techniques (conventional CT, MRI) and which is suitable for accurate repeated measurements
- Tumor sample confirmed as KRAS or BRAF mutation positive
- Patients must be able to swallow selumetinib capsules without regurgitation.

VII. Study Schedule

- Irinotecan d1, 15
- Selumetinib (50-75 mg po bid or 75 mg po qd)
- "Sparse" PK sampling day 1 and 15 (4 samples)
- Baseline tissue (PEFFT)
- LVEF if NYHA class 1 (and for new CHF)
- Ophthalmology evaluation if visual disturbance
- Heme, Chems, EKG, CT scans within 14 days
- Tumor biopsy baseline and d15 (±3d) optional

VIII. Dose finding portion

- Single agent dose of selumetinib 75 mg po bid
- Also tolerated in combination with docetaxel (phase I and NSCLC) and DTIC at 75 mg po bid
- Dose finding (3+3) design:

Dose Level	Dose of selumetinib – continuous (Day 1 through Day 28 - no planned interruptions)	Dose of irinotecan - every 14 days (on Day 1 and Day 15)
-1	75mg qd	iv 180 mg/m2
1	50 mg b.i.d.	iv 180 mg/m2
2	75 mg b.i.d.	iv 180 mg/m2

IX. Phase II Portion (Part B)

- Irinotecan 180 mg iv q 2 weeks with determined dose of selumetinib
- Primary endpoint PFS
- Secondary endpoints: RR, toxicity
- CT scan q 2 cycles (28 day cycles)
- 45 evaluable patients for PFS
 - 6 from part A plus 39 part B
 - Primary endpoint ORR (compared with 4% historical controls)
 - Early stopping at N=20 with ≤1 responses
 - Continue enrollment to 45; if ≥4 or more responses, then "promising regimen"
 - 10% probability of incorrectly determining ORR>4% (α= 0.10)
 - 90% probability of determining active regimen if true RR>15% (ß= 0.10)

X. Translational endpoints

- PEFFT baseline tissue
- Subset of patients with biopsy of accessible lesions on day 15 (±3)
- At least 15 paired specimens
- Endpoints
 - Total and activated ERK
 - Total and activated EGFR
 - TOPO I
 - Ki-67, p27, TUNEL
 - Array profile
- To be funded by AGICC
- R21 grant application pending (Eckhardt)

XI. RESULTS

- Enrolled = 3 patients (DL 1)
- Demographics
 - Male = 2, Female = 1
 - Caucasian = 3
- Cycles completed: 4,4,2
- Toxicity
 - Thrombocytopenia gr 3 = 1
 - ANC gr 3 = 1
 - Diarrhea gr 1 = 3
 - No grade 4 toxicity
- RESPONSES : SD = 2, PR = 1 (confirmed)

XII. CONCLUSIONS

- No unexpected toxicities seen
- No DLTS at DL 1
- Accrual continues at DL2
 - Irinotecan 180 mg/m2 q2w with selumetinib = 75 po bid
- 1 confirmed PR (partial response) (of 3 patients treated)
- We believe that a strategy of biomarker-based patient selection will maximize the opportunity for successful drug development through identifying the optimal clinical situation
 - In this case selecting tumors with KRAS or BRAF activating mutations to demonstrate a downstream signal transduction inhibitor can improve activity of standard chemotherapy

Supported by a grant from Astrazeneca