DNA Repair and Apoptosis

A phase II trial to assess the single-agent efficacy and safety of the PARP inhibitor olaparib (O) in previously treated patients with metastatic, measurable colorectal cancer (mCRC) stratified by microsatellite status (MSs).

L. P. Leichman[1], S. J. Cohen[2], H. S. Hochster[3], W. Messersmith[4], H. Lenz[5], B. M. Boman[6], P. J. Gold[7], B. H. O'Neil[8], J. Berlin[9], J. Carmichael[10]

[1]Comprehensive Cancer Center at Desert Regional Medical Center; [2]Fox Chase Cancer Center; [3]Yale Cancer Center; [4]University of Colorado Cancer Center; [5]University of Southern California, Los Angeles, CA; [6]Helen F. Graham Cancer Center; [7]Swedish Cancer Institute; [8]University of North Carolina at Chapel Hill; [9]Vanderbilt-Ingram Cancer Center; [10]AstraZeneca, Macclesfield, UK

Abst. ID: 118 (Temp. Abst. ID: 60883)

A phase II trial to assess the single-agent efficacy and safety of the PARP inhibitor olaparib (O) in previously treated patients with metastatic, measurable colorectal cancer (mCRC) stratified by microsatellite status (MSs).

DNA Repair and Apoptosis

Background: Polyadenosine 5'diphosphoribose polymerization (PARP) is a post-translational process contributing to cell survival after DNA damage by binding to single and double-stranded DNA breaks. The inability to repair damaged DNA is the leading cause of cancer for patients with BRCA1 and BRCA2 mutations (muts). O, is active against breast and ovarian cancers in patients with BRCA1 and BRCA2 muts. Similar susceptibility may exist for DNA instability associated with MSI-H mCRC. Therefore, AGICC initiated a phase II trial to test O against mCRC stratified for MSs. **Methods:** Patients with mCRC failing standard tx and ECOG PS 0-1 were eligible regardless of MSs. Patients received O 400 mg po bid x 28 days (one cycle). MSs was performed centrally in a CLIA certified lab. CT scan evaluations were q 8 wks. Using a Simon two-stage design for each stratum: if ≥ 3/17 responses in non-MSI-H patients, enrollment for this cohort would continue to 29. If < 3/17 responses in the first 17 patients, eligibility would be restricted to a cohort of 15 MSI-H patients. MSs is confirmed centrally in a CLIA certified lab by RT-PCR. Pre-tx slides were submitted for PARP evaluation; within 7 to 10 days of tx an additional optional biopsy may be obtained to determine the effect of O on intratumoral PARP. **Results**: 20 non-MSI-H patients were enrolled in 3 months. Demographics: median (m) age: 56 (range 39-82); m PS: 1. M cycles of O: 2. 4 patients had either grade III/ IV non-heme toxicities, fatigue most common. No responses were reported in non MSI-H cohort. Biopsies while on O have been obtained from 7 patients. 10 MSI-H patients have been enrolled; response data for MSI-H patients are not yet available. **Conclusions:** O was well tolerated but has no single-agent activity against non-MSI-H mCRC. Accrual of MSI-H mCRC patients continues. Responses for MSI-H patients will be reported as will the changes in intra-tumoral PARP levels. MSI-H patients with mCRC represent a small fraction of the total mCRC population (10 patients enrolled, >150 patients screened). However, finding active agents for MSI-H patients may have relevance to a greater number in the adjuvant setting.

Authors' Disclosures

- Lawrence Leichman, MD — Consultant or Advisory Role, AstraZeneca; Honoraria, AstraZeneca; Research Funding, AstraZeneca
- Steven Cohen, MD — Research Funding, AstraZeneca
- Howard Hochster, MD — Consultant or Advisory Role, AstraZeneca; Research Funding, AstraZeneca
- Wells Messersmith, MD — Consultant or Advisory RoleAstraZeneca; Research FundingAstraZeneca
- Heinz-Josef Lenz, MD — Consultant or Advisory Role, AstraZeneca; Research Funding, AstraZeneca
- Bruce Boman, MD, PhD — Research Funding, AstraZeneca
- Philip Gold, MD — Research Funding, AstraZeneca
- Bert O'Neil, MD — Research Funding, AstraZeneca
- Jordan Berlin, MD — Consultant or Advisory Role, AstraZeneca; Research Funding, AstraZeneca
- James Carmichael, MD — Employment/Leadership Position, AstraZeneca; Stock Ownership, AstraZeneca

Aptium Oncology GI Cancer Consortium

I. Background

Polyadenosine 5'diphosphoribose polymerization (PARP) is a post-translational process contributing to cell survival after DNA damage by binding to single and double-stranded DNA breaks. The inability to repair damaged DNA is the leading cause of cancer for patients with BRCA1 and BRCA2 mutations (muts). Olaparib, is active against breast and ovarian cancers in patients with BRCA1 and BRCA2 muts. Similar susceptibility may exist for DNA instability associated with MSI-H mCRC. Therefore, AGICC initiated a phase II trial to test olaparib against mCRC stratified for MSs.

MMR Defects

- **HNPCC:** represent between 3-5% of all CRC patients. These patients exhibit germ-line mutations within specific repair genes leading to MSI-H phenotype.
- **Sporadic**: represent ~15% of all CRC caused by methylation defects (epigenetic event) leading to the MSI-H patients phenotype.
- These defects result in the accumulation of abnormalities of short sequences of nucleotide bases or "microsatellites" that are repeated 10-100s of times in the genome of the tumor (somatic) or entire host (germline).

Molecular Mechanisms Leading to CRC
80% CIN; 20% epigenetic silencing or MSI

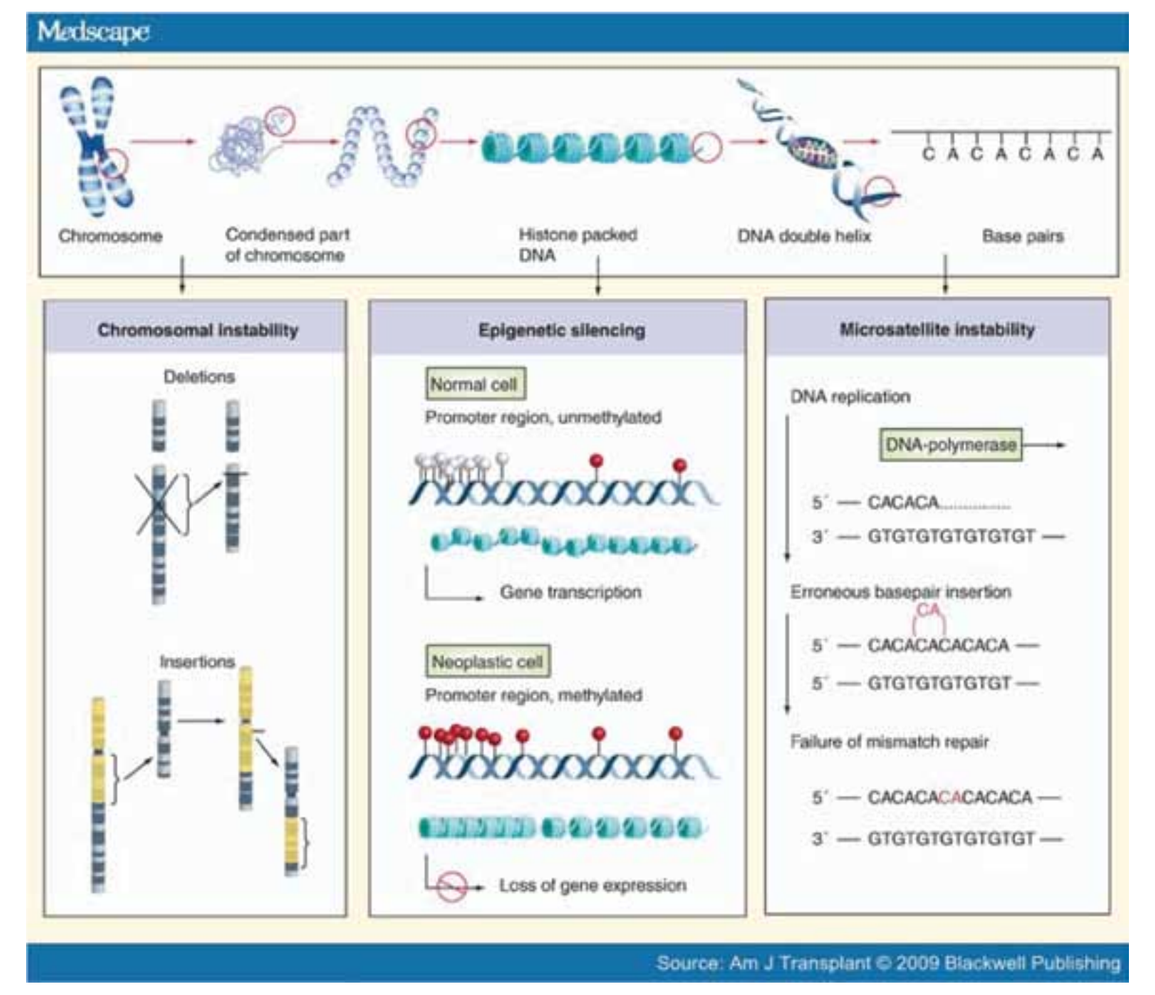

Medscape
Chromosome
Condensed part of chromosome
Histone packed DNA
DNA double helix
Base pairs
Chromosomal instability
Deletions
Insertions
Epigenetic silencing
Normal cell
Promoter region, unmethylated
Gene transcription
Neoplastic cell
Promoter region, methylated
Loss of gene expression
Microsatellite instability
DNA replication
DNA-polymerase
Erroneous basepair insertion
Failure of mismatch repair
Source: Am J Transplant © 2009 Blackwell Publishing

PARP and PARP Inhibition

- Polyadenosine 5'-diphosphoribose [poly-(ADP-ribose)] or PAR polymerisation is a unique post-translational modification of histones and other nuclear proteins that contributes to the survival of proliferating and non-proliferating cells following deoxyribonucleic acid (DNA) damage.

PARP FUNCTIONS

- The range of biological roles involving PARP proteins is wide including DNA repair and maintenance of genomic integrity.

PARP-1 and DNA Repair

A model showing DSB formation and DNA rearrangement in the absence of BRCA and PARP1. (A) PARP-1 binds and repairs single strand breaks (SSBs) in DNA. Unrepaired SSBs result in formation of double strand breaks (DSBs) when they meet replication forks. (B) In the presence of wild type BRCA1 and BRCA2, DSBs can be efficiently repaired by RAD51 mediated homologous recombination (HR). (C) BRCA1 or BRCA2 mutant cells cannot repair DNA DSBs properly upon the inhibition of PARP, leading to DNA replication arrest and illegitimated DNA ends joining.

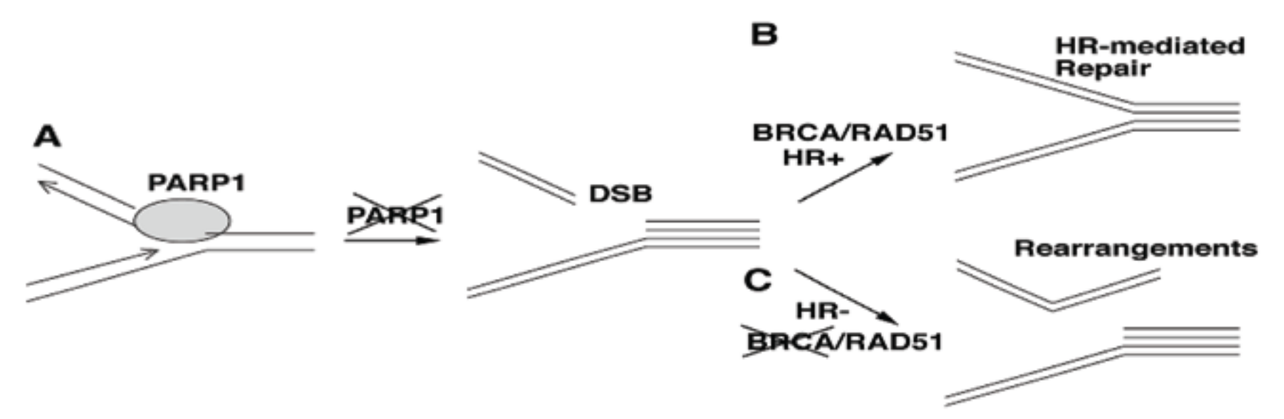

A
PARP1
~~PARP1~~
DSB
B
BRCA/RAD51
HR+
HR-mediated Repair
C
HR-
~~BRCA~~/RAD51
Rearrangements

Olaparib

Olaparib is an inhibitor of PARP (poly-ADP ribose polymerase). As such, the drug may block the DNA repair pathway known as Base Excision Repair (BER) resulting in accumulation of unrepaired DNA single strand breaks (SSBs). During DNA replication, these unrepaired SSBs are converted into potentially cytotoxic DNA double strand breaks (DSBs).

Rationale for olaparib as a single agent against CRC

Although a PARP inhibitor may be best utilized with a DNA damaging agents such as oxaliplatin or irinotecan, safety data for olaparib in combination with oxaliplatin or irinotecan are not yet available.

Single agent activity in either the MSI-H or non MSI-H groups would be a great impetus to study olaparib earlier in the disease process.

II. Hypotheses

1. Olaparib administered to patients with disseminated CRC having progressed during or after standard systemic therapy will result in clinical benefit (partial or complete responses) and disease stabilization (progression free survival).
2. Patients who exhibit germ line mutations in mismatch repair genes (MSI-H) or those patients with sporadic CRC with evidence of epigenetic silencing of mismatch repair genes (MSI-H) will have higher response rates and longer progression-free survival (PFS) than those patients whose tumors are non-MSI-H.

Exploratory Translational Objectives

1. To determine whether PARP-1 levels found in archival tissue prior to treatment have a relationship to response and/or toxicity for patients treated with olaparib.
2. In patients consenting to biopsy during therapy, to measure and explain drug activity levels after 7 to 10 days of therapy with olaparib.
3. To determine if pharmacogenomic parameters as determined from pre-therapy blood samples affect response or toxicity to olaparib.

III. Clinical Methodology - Trial 09CRC01

- Patients with mCRC failing standard tx and ECOG PS 0-1 eligible regardless of MS status
- Patients receive olaparib 400 mg po bid; one cycle was 28 days
- MS status was performed centrally in a CLIA certified lab by RT-PCR under the direction of Dr. Stanley Hamilton at the M.D. Anderson Cancer Center.
- CT scan evaluations q 8 wks

Olaparib:

Dose, Route/Schedule of Administration

- Patients self-administered olaparib 400 mg po bid continuously with interruption only for grade 3 or grade 4 toxicity (see below).
- The dose was NOT to be adjusted according to the patient's body weight. Dose intervals were as close to every twelve hours each day as possible.
- Using a Simon two-stage design for each stratum, if ≥ 3/17 responses in non-MSI-H patients, enrollment for this cohort would continue to 29. If < 3/17 responses in the first 17 patients, eligibility would be restricted to a cohort of 15 MSI-H patients.
- Pre-tx slides submitted for PARP evaluation; within 7 to 10 days of tx an additional optional biopsy may be obtained to determine the effect of olaparib on intratumoral PARP.
- Pre-therapy serum for genomic markers all patients.

IV. Current Results

- 20 non-MSI-H patients were enrolled in 3 mos
- Median age: 56 (range 39-82)
- Median PS: 1
- Median # cycles of olaparib: 2
- 4 patients had either grade 3 or 4 non-heme toxicities, fatigue most common
- 8 biopsies obtained while on olaparib to compare with base-line tumor
- 10 MSI-H patients have been enrolled
- Response data for MSI-H patients are not yet available

V. Conclusions

Based on preliminary data, olaparib was well tolerated but has no single-agent activity against non-MSI-H mCRC. Accrual of MSI-H mCRC patients continues. Responses for MSI-H patients will be reported as will the changes in intratumoral PARP levels. MSI-H patients with mCRC represent a small fraction of the total mCRC population (10 patients enrolled, >150 patients screened). However, finding active agents for MSI-H patients may have relevance to a greater number in the adjuvant setting.

- Olaparib was well tolerated in both non-MSI-H and MSI-H patients
- Rapid accrual of non-MSI-H mCRC patients to 3rd line therapy attests to need for new agents in CRC
- MSI-H patient accrual continues
- MSI-H patients with disseminated CRC who fail to standard therapy represent less than 3% of all patients eligible for this protocol

Although it may not be feasible to develop agents targeting MSI-H patients with disseminated CRC (3% or less), a positive outcome could have greater meaning in the adjuvant setting where closer to 20% of patients are MSI-H.

Even in the 3rd line setting, highly motivated patients are willing to undergo biopsies for purposes of hypothesis generating data if the rationale for the biopsies are explained by highly motivated investigators.

Efficacy results and the results of the biopsies for patients on this trial will be the subject of a separate report.

Supported by a grant from Astrazeneca